RECOVERY IS *REAL*

HOPE, CHOICE, AND COLLABORATION ARE *ESSENTIAL.*

From the APHA Policy Statement: *Support for Social Determinants of Behavioral Health and Pathways for Integrated and Better Public Health, November 18, 2014*

Behavioral health is an essential component of overall health and well-being.

Research on the social determinants of behavioral health, along with increasing acknowledgment of the evidence by policymakers, provides a basis for public health strategies to improve the nation's behavioral health.

Recovery-oriented care is not a new model, it is about fundamentally doing differently what we do every day

A 2014 report from the American Academy of Family Physicians noted that one-half of primary care patients have a behavioral condition that is significantly disabling. Behavioral health is integral to overall health.[1]

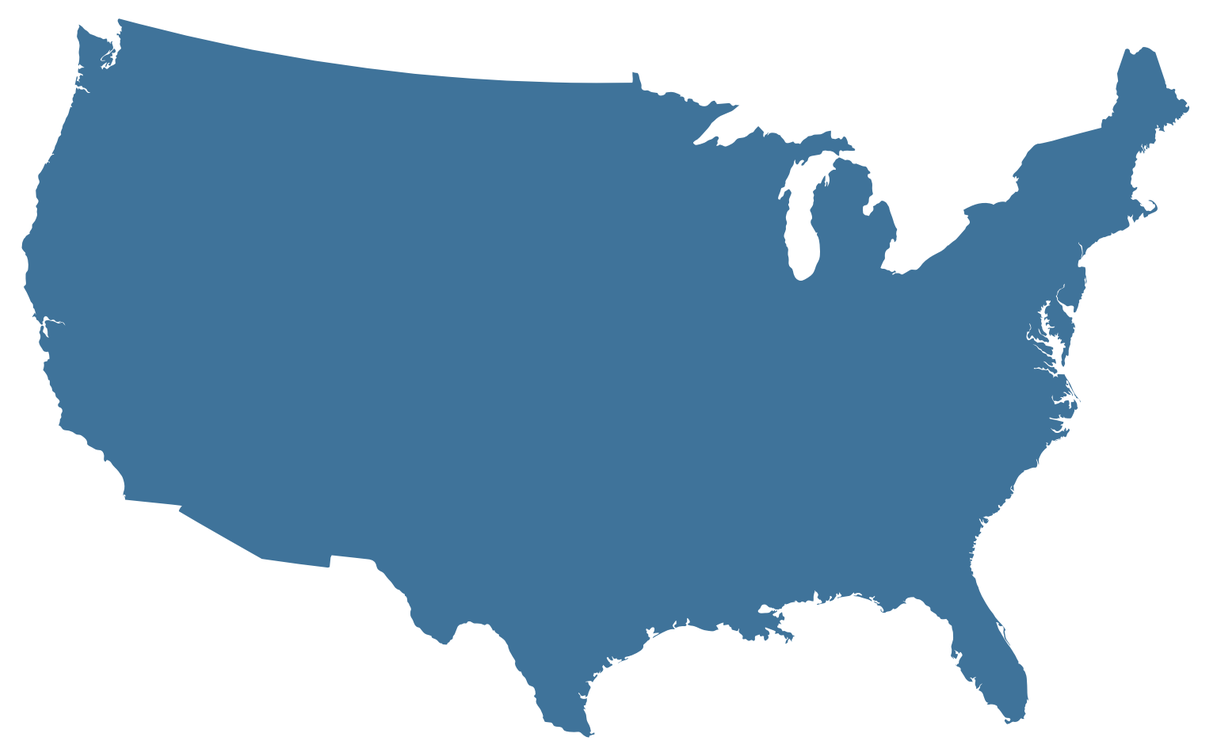

People with serious mental health conditions die earlier than people without these conditions. The leading cause of the decreased longevity was not the behavioral health condition, but common and often preventable conditions, particularly cardiovascular, pulmonary, and metabolic disorders.[2]

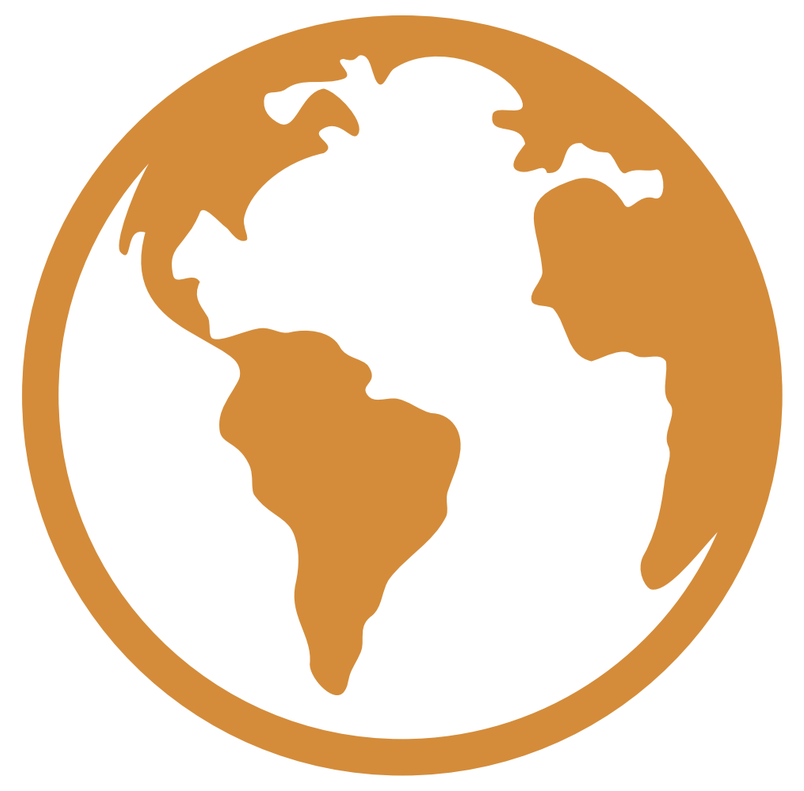

A 2006 U.S. study found similar results and emphasized the critical impact of social determinates of behavioral health on this reduced longevity: poverty, homelessness, unemployment, side effects of psychotropic medications, social isolation, trauma and discrimination.[3]

What *YOU* can do:

- Use RTP to help you build workforce knowledge and skills in recovery-oriented practices that have been shown to be effective in helping people recover their full potential
- Promote positive social determinants of health and recovery
- Act to end discrimination and pessimism in behavioral health prognosis
- Spot the impact of trauma: emotional, physical, environmental, refugee
- Use welcoming approaches and model inclusion
- Attend to choice and self-determination
- Collaborate as partners in care; use person-centered planning
- Promote and support self-care activities
- Practice shared decision making
- Respect and value peer-delivered services
- Address social determinates of health drawing from SAMHSA evidence based practices KITS: http://store.samhsa.gov/list/series?name=Evidence-Based-Practices-KITs

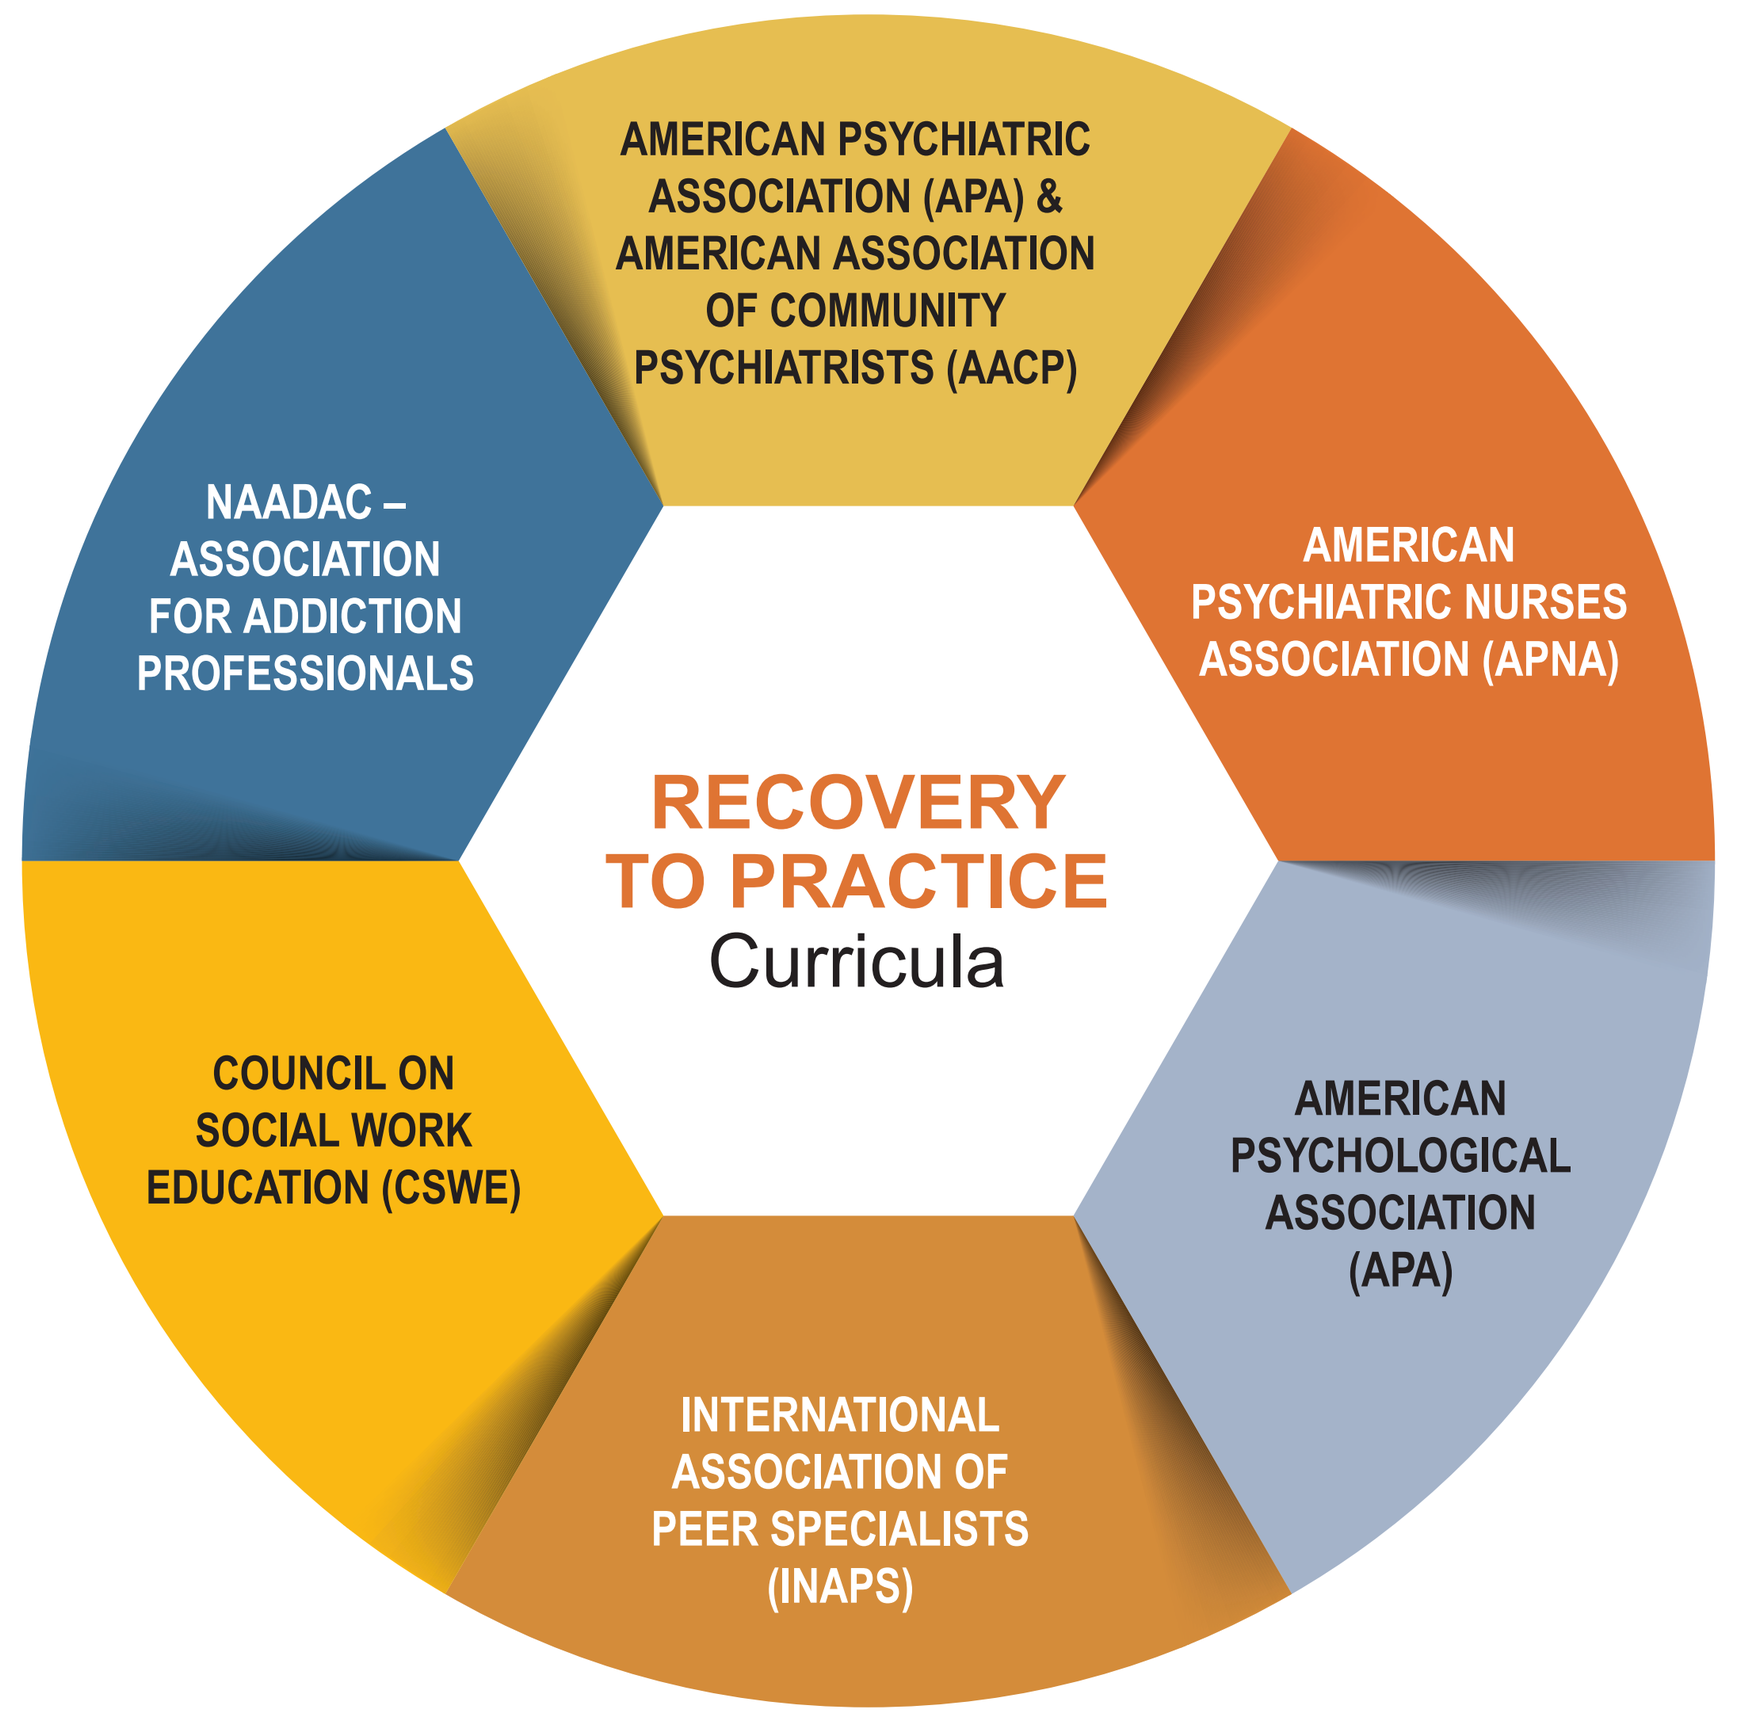
AMERICAN PSYCHIATRIC ASSOCIATION (APA) & AMERICAN ASSOCIATION OF COMMUNITY PSYCHIATRISTS (AACP)
AMERICAN PSYCHIATRIC NURSES ASSOCIATION (APNA)
AMERICAN PSYCHOLOGICAL ASSOCIATION (APA)
INTERNATIONAL ASSOCIATION OF PEER SPECIALISTS (INAPS)
COUNCIL ON SOCIAL WORK EDUCATION (CSWE)
NAADAC – ASSOCIATION FOR ADDICTION PROFESSIONALS
RECOVERY TO PRACTICE
Curricula

- *Recovery to Practice [RTP] offers 6 discipline-specific curriculum to help build the recovery practice skills of your integrated and multidisciplinary workforce. These curriculum are supplemented by virtual training events and technical assistance.*

PEOPLE RECOVER FROM BEHAVIORAL HEALTH CONDITIONS. *HOW?*

HEALTH
- Access to Treatment and Services
- Welcome
- Attention to preventable causes of illness
- Engagement/Activation
- Health Literacy

HOME
- Housing
- Safety
- Clean Environment
- Nutrition

COMMUNITY
- Social and community networks
- Inclusion
- Social Support
- Positive role models

PURPOSE
- Self Determination
- Work/Economic security
- Education
- Spirituality
- Creative expression
- Intellectual pursuits

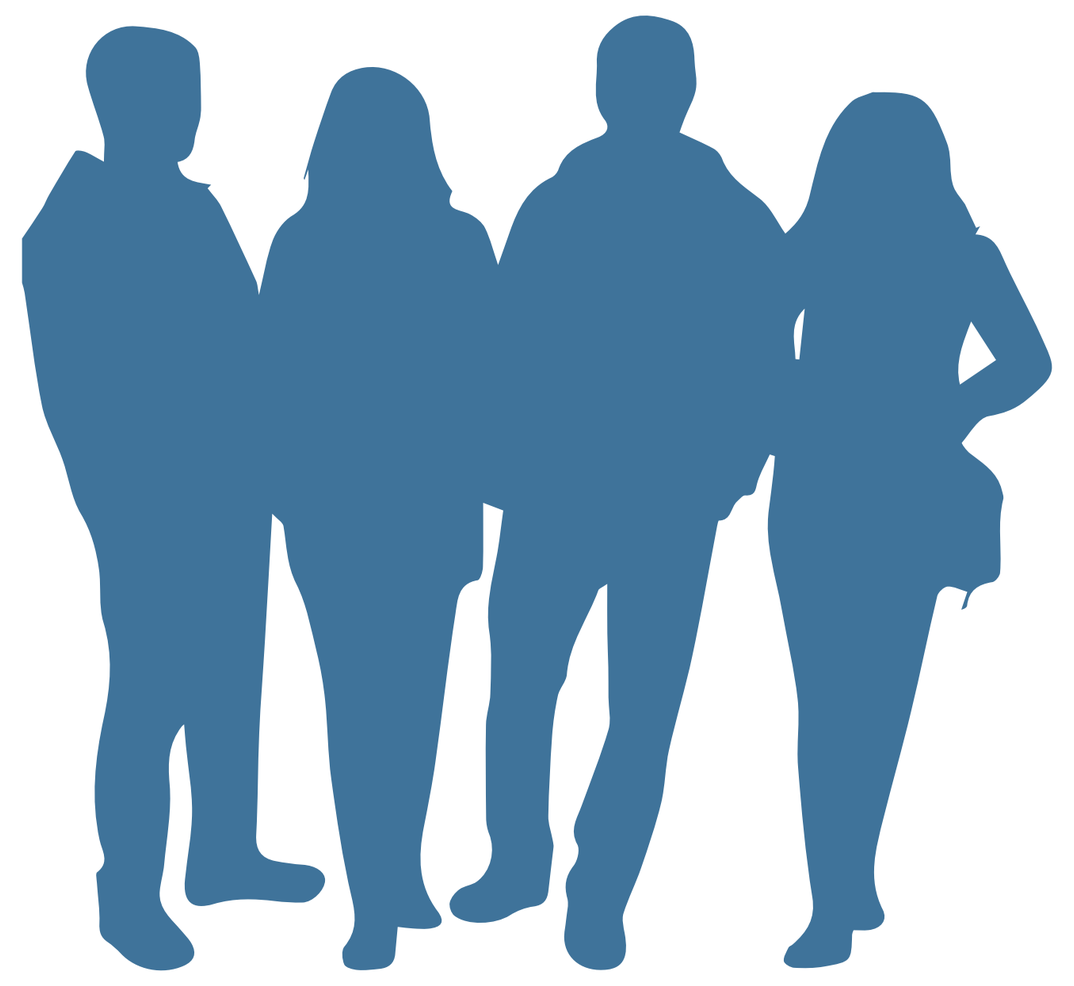

Social Determinants are the cultural, social, economic, health, and environmental conditions at the national, regional, community, and family levels that influence one's health, wellness, and recovery from physical and behavioral health conditions.

Recovery to Practice
Resources for Behavioral Health Professionals

Substance Abuse and Mental Health Services Administration
SAMHSA
www.samhsa.gov • 1-877-SAMHSA-7 (1-877-726-4727)

[1] *Source: The Working Party Group on Integrated Behavioral Healthcare:, Baird, M., Blount, A., Brungardt, S., Dickinson, P., Dietrich, A., … deGruy, F. (2014). Joint Principles: Integrating Behavioral Health Care Into The Patient-centered Medical Home. Annals of Family Medicine, 12(2), 183–185. http://doi.org/10.1370/afm.1633*

[2] *Walker ER, McGee RE, Druss BG. Mortality in Mental Disorders and Global Disease Burden Implications: A Systematic Review and Meta-analysis. JAMA Psychiatry. 2015 Feb 11. doi: 10.1001/jamapsychiatry.2014.2502.*

[3] *Parks J., Svendsen D., Singer P. and Foti M. (2006) Morbidity and Mortality in People with Serious Mental Illness. National Association of State Mental Health Program Directors (NASMHPD) Medical Directors Council, Alexandria, VA 22314. Available at: www.nasmhpd.org/general_files/publications/med_directors_pubs/Technical%20Report%20on%20Morbidity%20and%20Mortaility%20-%20Final%2011-06.pdf(accessed17March2010).*

www.samhsa.gov/recovery-to-practice